A Phase 2 Study of PCSK9 Inhibitor, Alirocumab, and PD1 Inhibitor, Cemiplimab, in Advanced Immunorefractory Metastatic NSCLC

E. Oduah[1,2], N. Ready[1,2], J. Clarke[1,2], J. Rivera Concepcion[1,2], J. Crawford[1,2], L. Alder[1,2], J. Gray[3], T. Stinchcombe[1,2], S. Antonia[1,2]
Affiliation: [1]Duke University, Durham/NC/USA ,[2]Duke Cancer Institute, Durham/NC/USA ,[3]Moffitt Cancer Center, Tampa/FL/USA
Correspondence: *Eziafa.oduah@duke.edu*

IASLC 2025 World Conference on Lung Cancer
SEPTEMBER 6-9, 2025
BARCELONA, SPAIN

INTRODUCTION

- Immunotherapy resistance is an unmet clinical need in NSCLC.
- PCSK9 is an immuno-inhibitory therapeutic target in preclinical models.
- First-in-class proof of principle clinical trial evaluating the clinical activity and safety of anti-PCSK9 (alirocumab) plus anti-PD1 (cemiplimab) in immunorefractory metastatic NSCLC.
- Preliminary efficacy findings in the first 40/60 patients are reported.

METHODS

Design:

- Phase 2, multi-institutional single arm trial.
- Sites:
 - Duke Cancer Institute
 - Moffitt Cancer Center
- Evaluate the clinical activity and safety of alirocumab (PCSK9 inhibitor) plus cemiplimab (anti-PD1 antibody) in metastatic NSCLC who had progressed on prior immune checkpoint blockade.
- Primary endpoint: ORR
- Secondary efficacy endpoints:
 - PFS, OS, DOR, DCR
- Correlative objectives:
 - Biomarkers of response
- Planned sample size: 60
- ClinicalTrials.gov ID NCT05553834
- Study approved by Duke IRB

RESULTS

Alirocumab + Cemiplimab demonstrated efficacy in immunorefractory NSCLC. In the evaluable patients (n= 38), ORR was 36.4% (4/11) in squamous and 3.7% (1/27) in non-squamous histology.

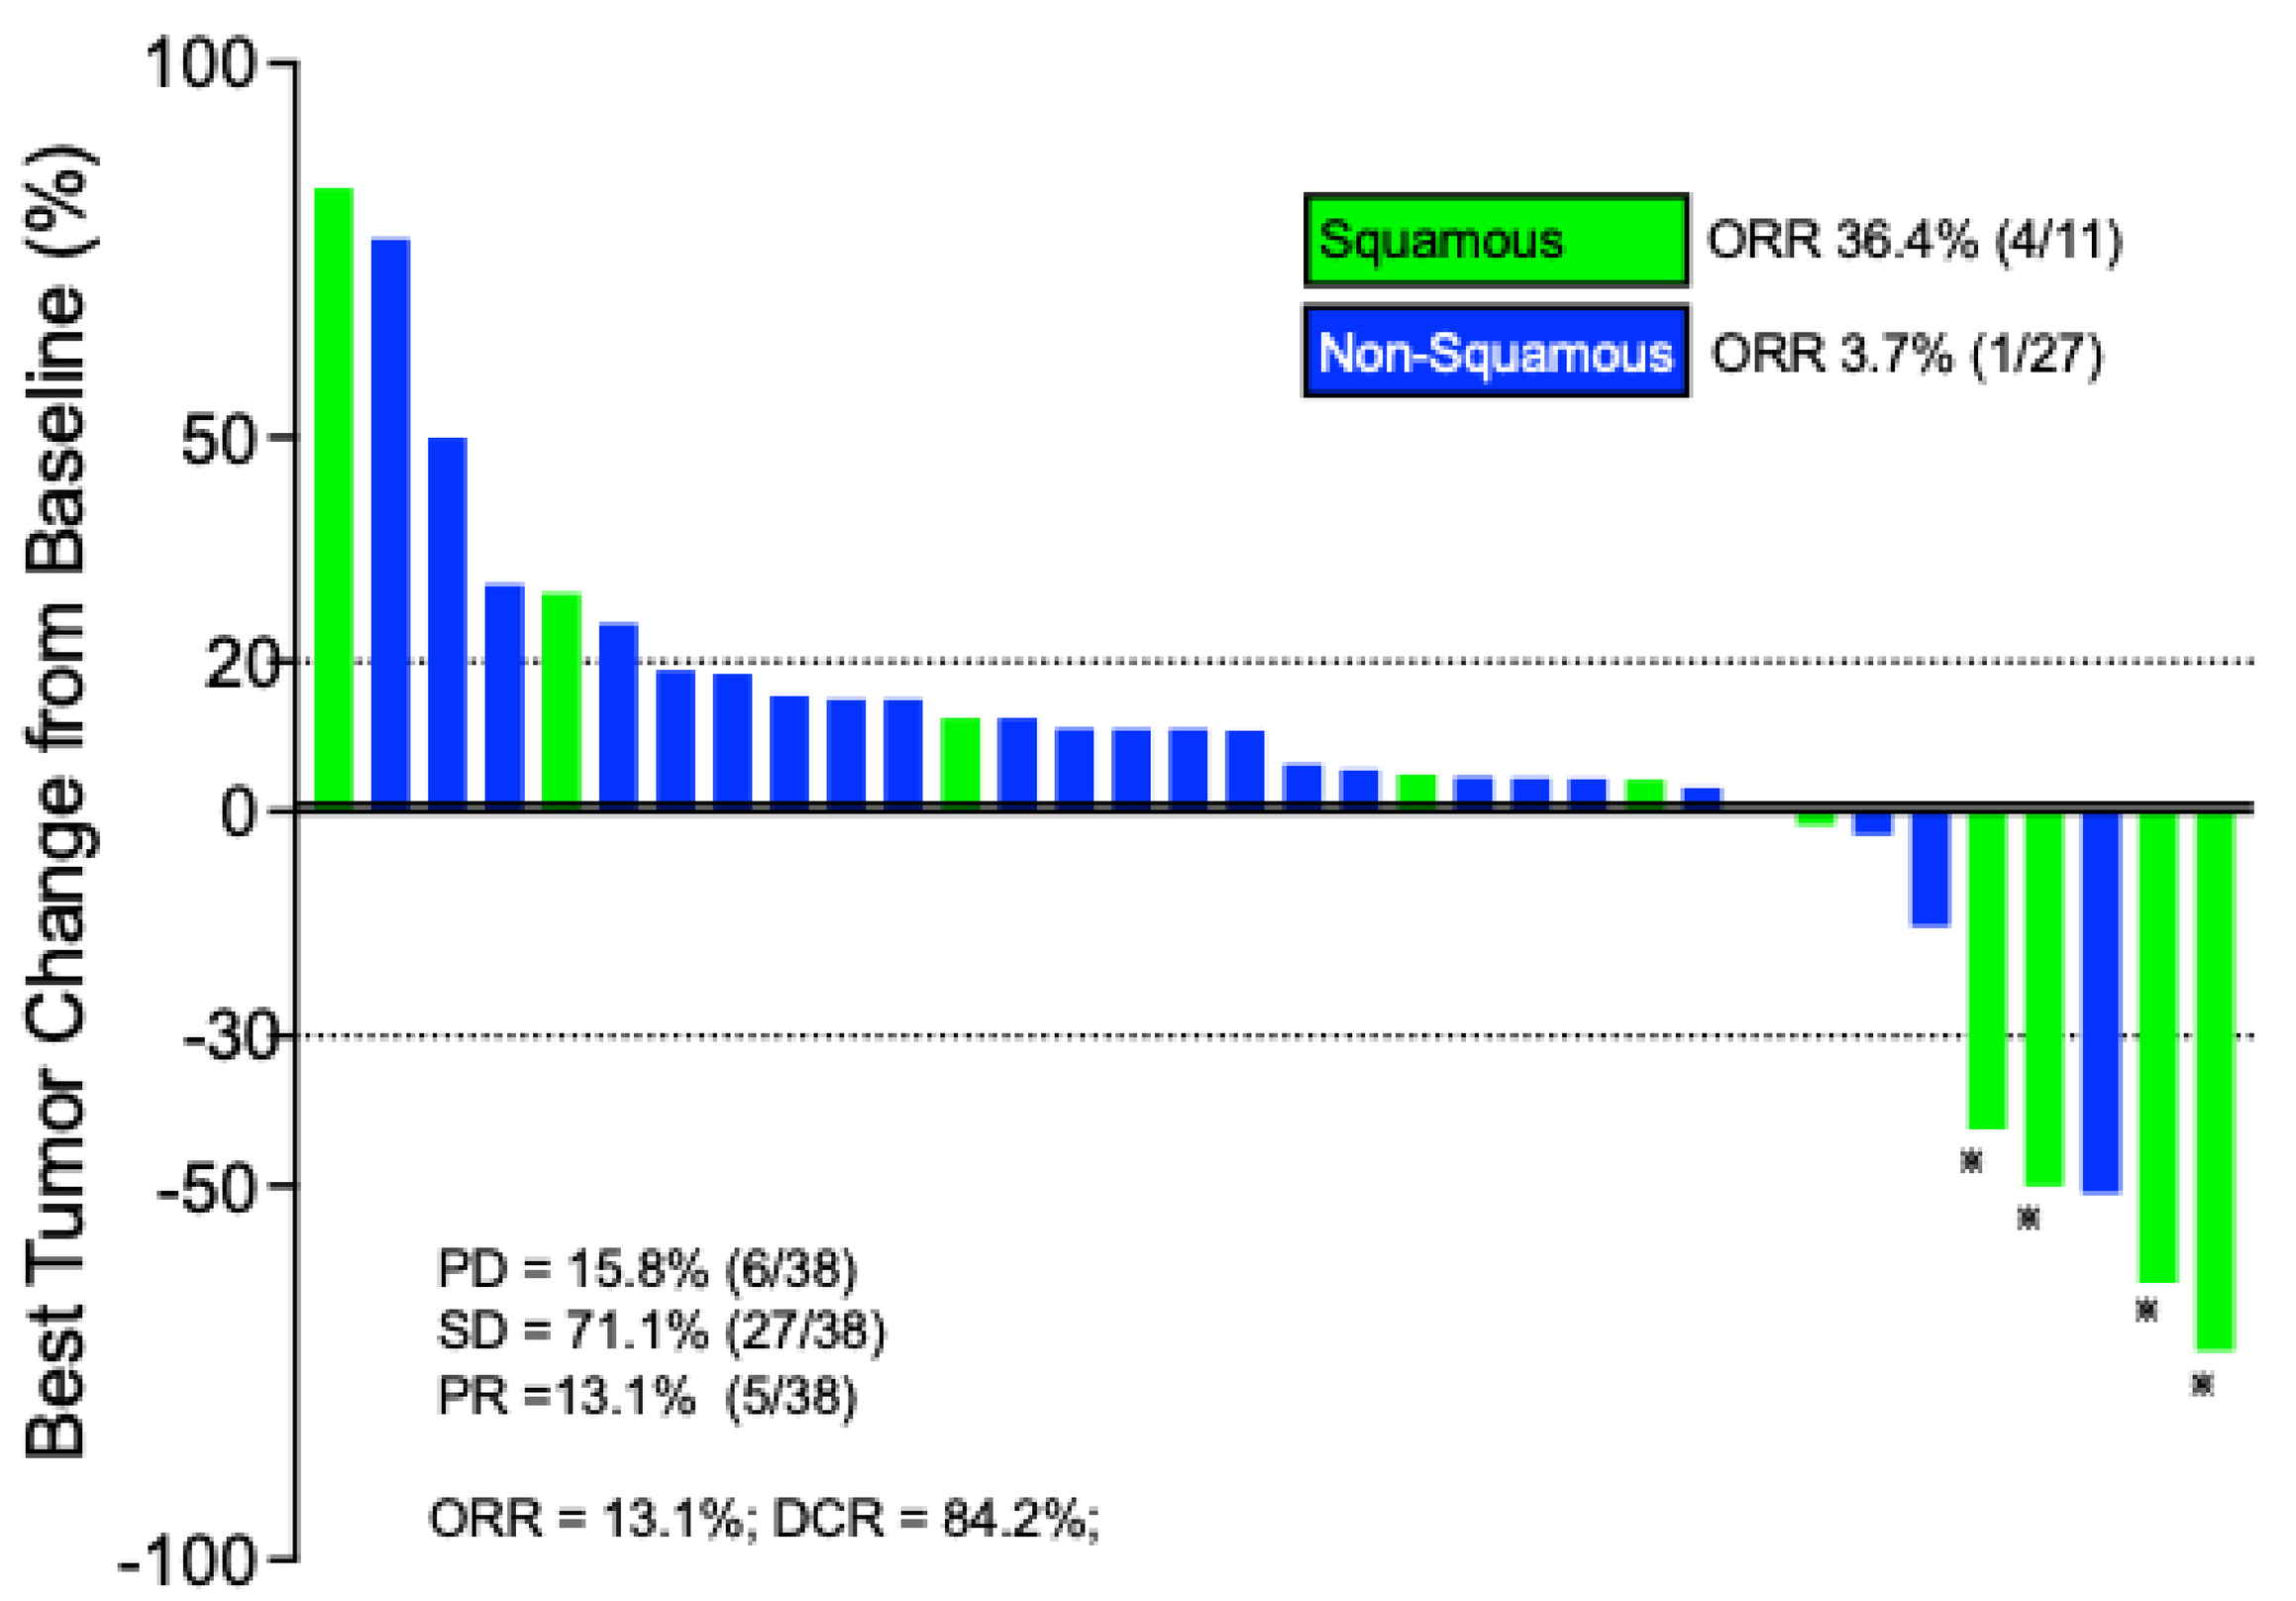
Best Tumor Change from Baseline (%)
100
50
20
0
-30
-50
-100
Squamous
ORR 36.4% (4/11)
Non-Squamous
ORR 3.7% (1/27)
PD = 15.8% (6/38)
SD = 71.1% (27/38)
PR =13.1% (5/38)
ORR = 13.1%; DCR = 84.2%;

Patients with squamous histology demonstrated statistically significant improved PFS and a trend towards improved OS.

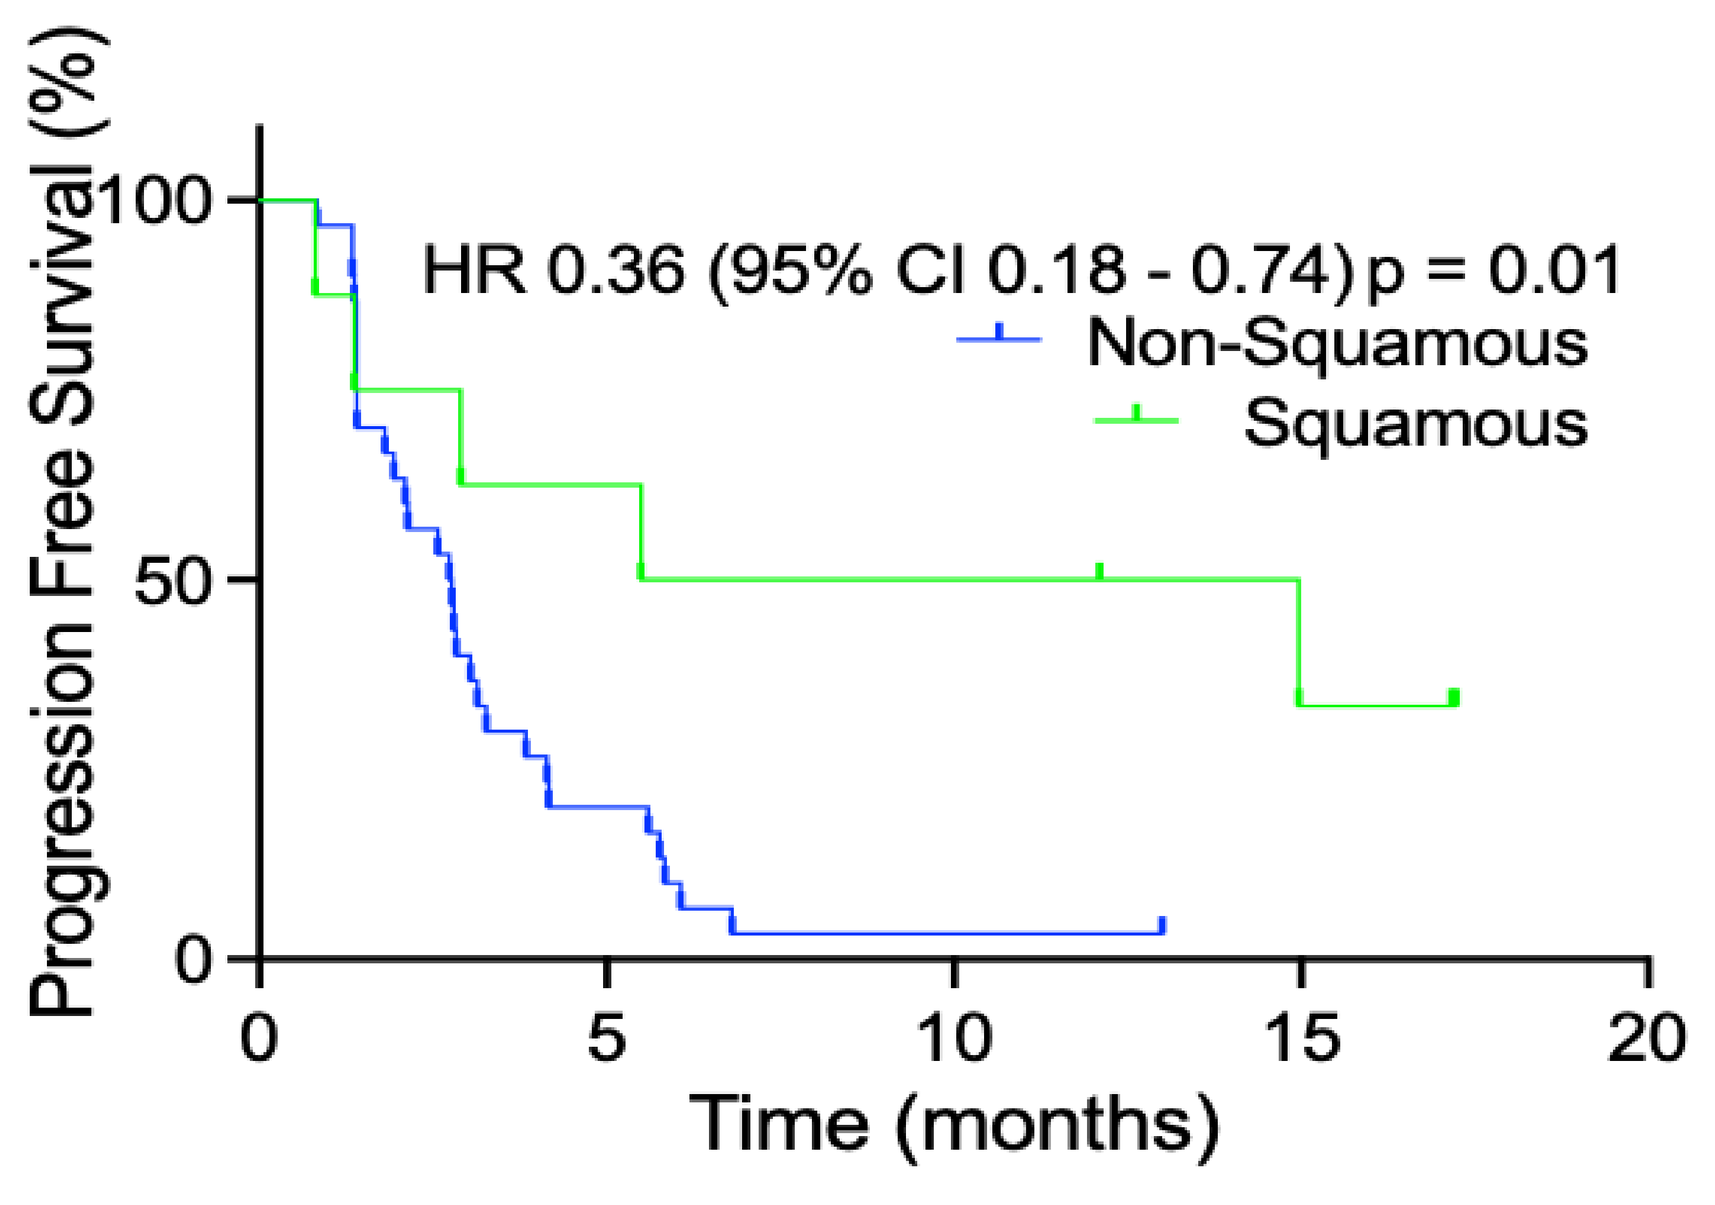
Progression Free Survival (%)
HR 0.36 (95% CI 0.18 - 0.74) p = 0.01
Non-Squamous
Squamous
Time (months)

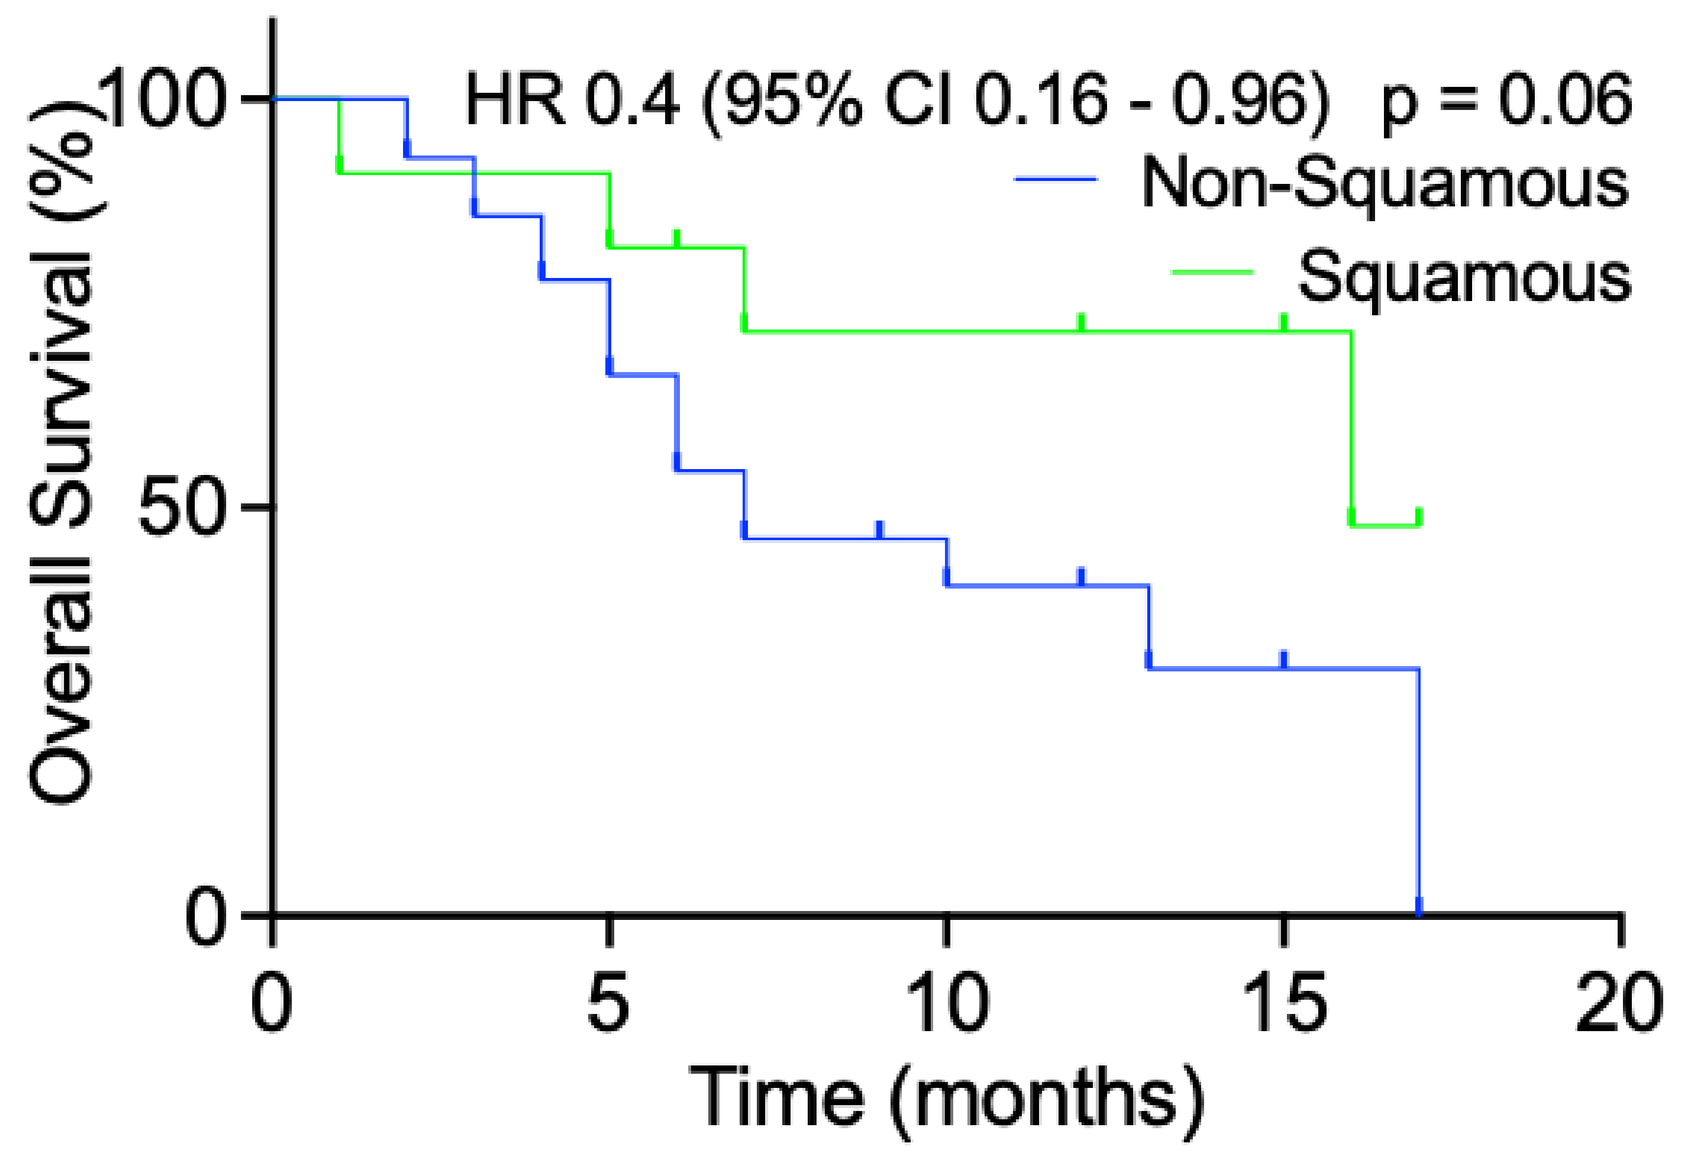
Overall Survival (%)
HR 0.4 (95% CI 0.16 - 0.96) p = 0.06
Non-Squamous
Squamous
Time (months)

All responders (5/5) had alterations in PIK3CA, AKT1 or PTEN and had durable responses. Altered (n = 10); Unaltered (n= 28)

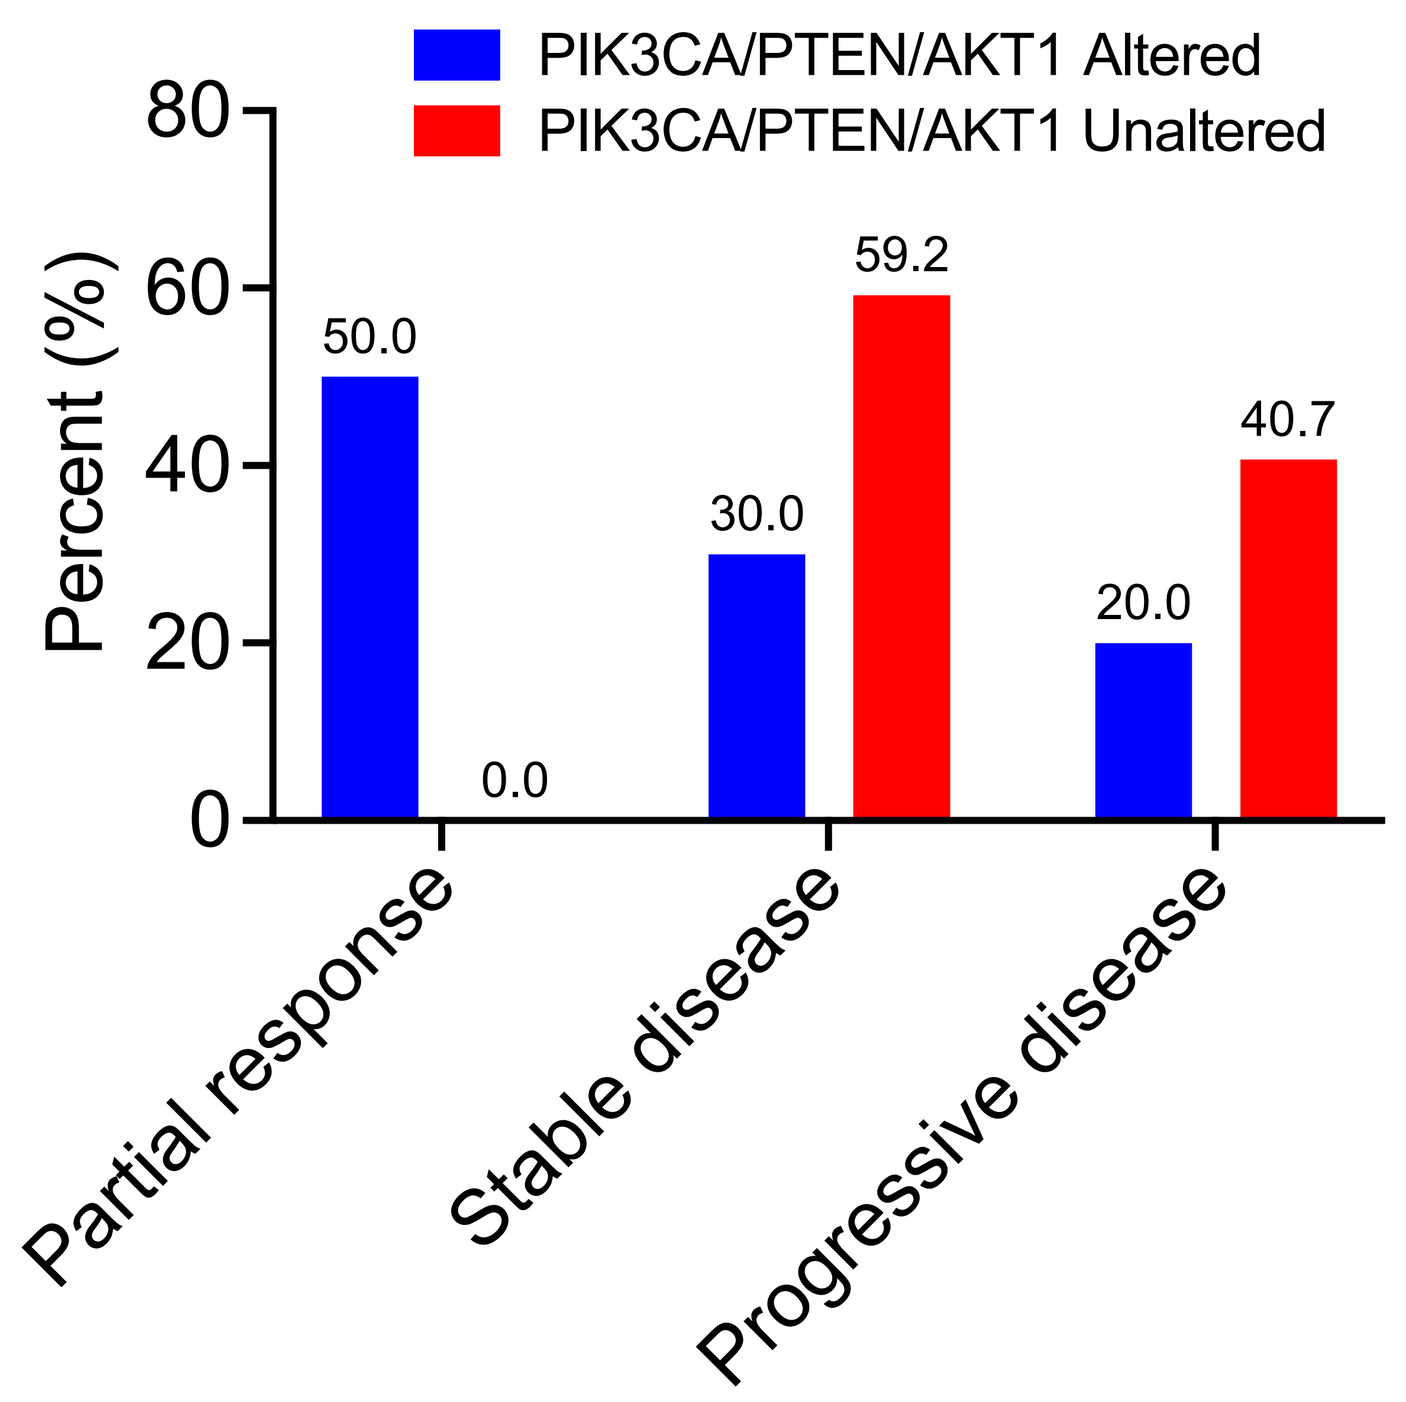
PIK3CA/PTEN/AKT1 Altered
PIK3CA/PTEN/AKT1 Unaltered
Percent (%)
50.0
0.0
30.0
59.2
20.0
40.7
Partial response
Stable disease
Progressive disease

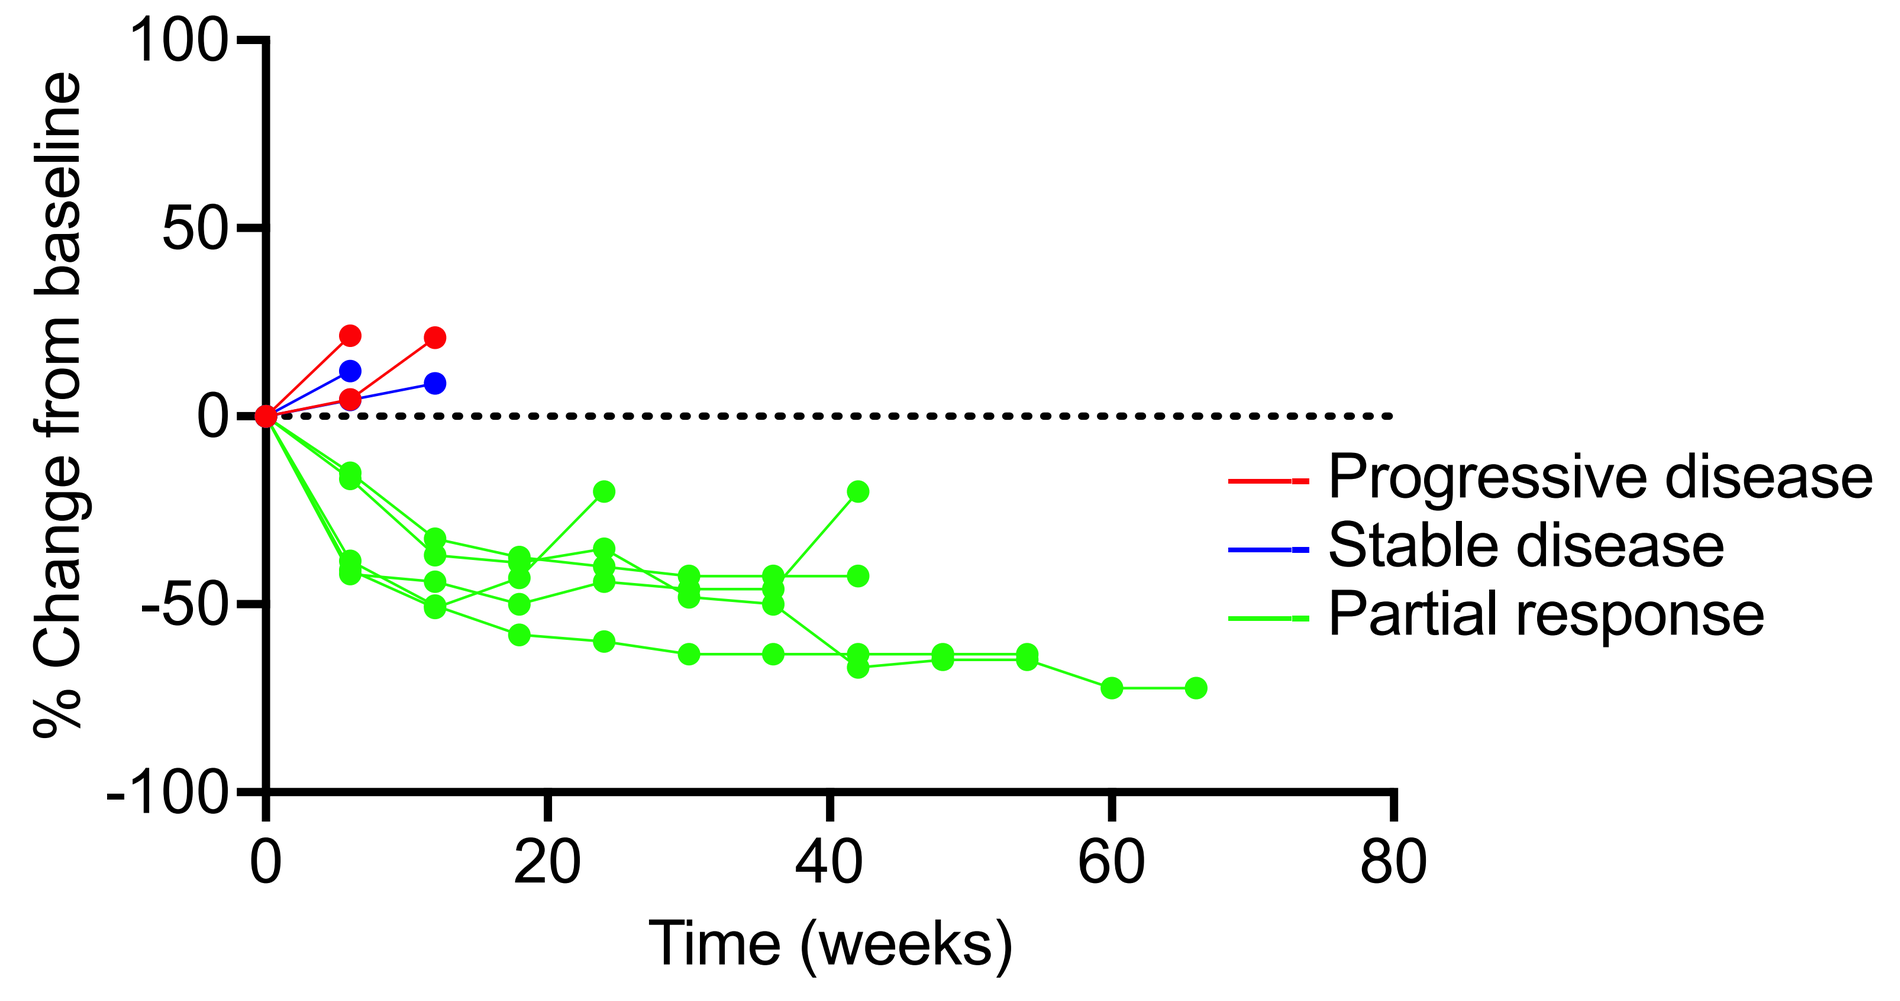
% Change from baseline
Progressive disease
Stable disease
Partial response
Time (weeks)

The presence of activating alterations in PIK3CA, AKT1 or PTEN predicted PFS (statistically significant) and OS (not statistically significant) benefit

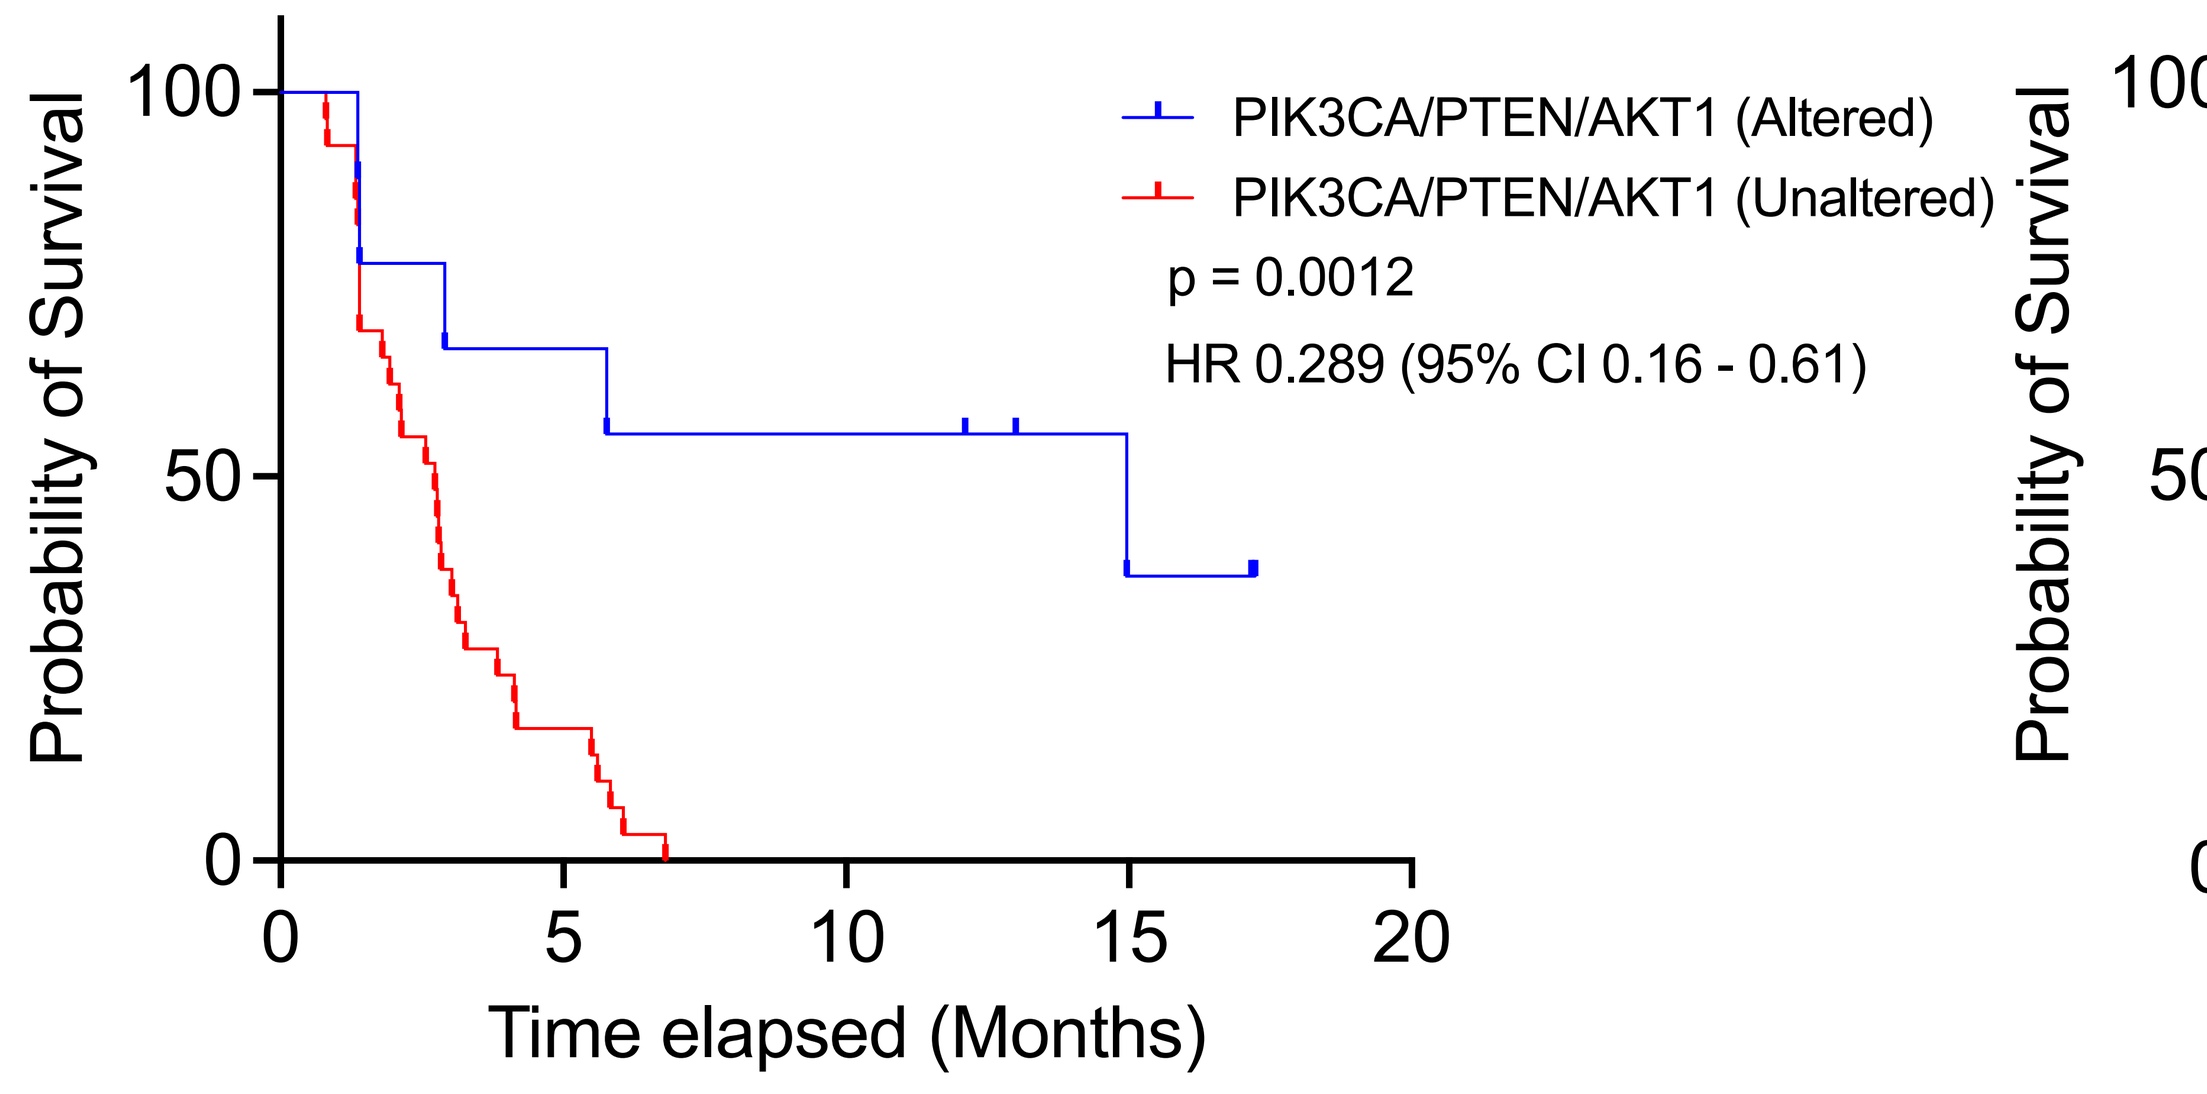
PFS by Alteration Status
Probability of Survival
PIK3CA/PTEN/AKT1 (Altered)
PIK3CA/PTEN/AKT1 (Unaltered)
p = 0.0012
HR 0.289 (95% CI 0.16 - 0.61)
Time elapsed (Months)

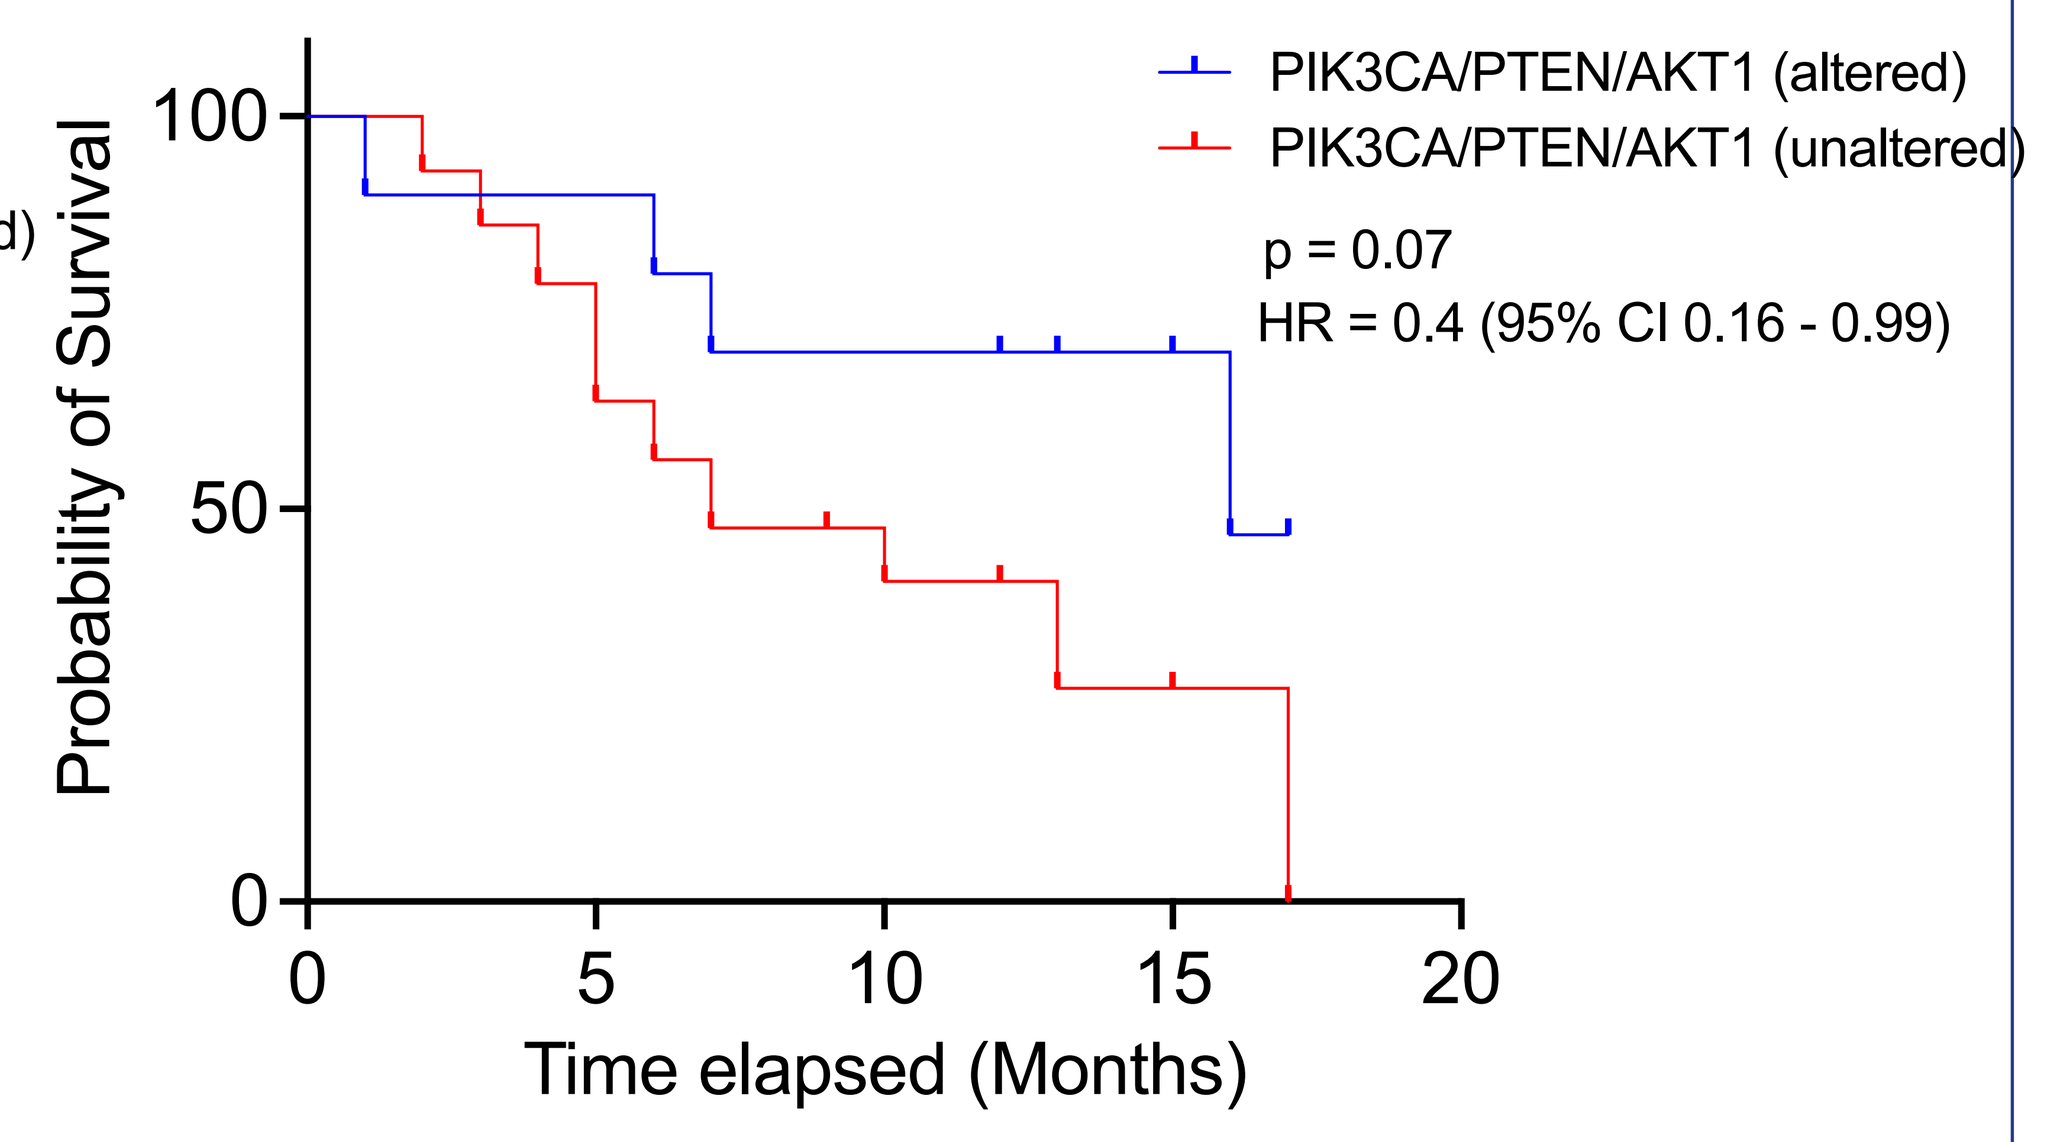
OS by Alteration Status
Probability of Survival
PIK3CA/PTEN/AKT1 (altered)
PIK3CA/PTEN/AKT1 (unaltered)
p = 0.07
HR = 0.4 (95% CI 0.16 - 0.99)
Time elapsed (Months)

Demographics and Clinical Characteristic	
Characteristic	**Population**
Median age (range), yr	67.0 (49-91)
Sex — no. (%)	
Male	19 (47.5)
Female	21 (52.5)
Race or ethnic group	
White	27 (67.5)
Black	12 (30)
Other	1 (2.5)
Smoking history — no. (%)	
Never	4 (10)
Current	2 (5)
Former	28 (70)
Unknown	5 (12.5)
ECOG performance-status grade — no. (%)	
0	1 (2.5)
1	39 (97.5)

Histology	
Squamous	11 (27.5)
Nonsquamous	29 (72.5)
Metastatic disease stage, no. (%)	
No. of previous lines of therapy, no. (%)	
1	13 (32.5)
2	14 (35)
3	4 (10)
>3	7 (17.5)
Unknown	2 (5)
Median prior #of therapy (r	2.0 (1–5)

Sponsor: Duke University
Funding: Regeneron

DISCUSSION & FUTURE DIRECTION

- No unexpected adverse events.
- Final analysis of outcomes is pending data maturation.
- Further investigation in a larger confirmatory trial is planned to confirm the trends in squamous NSCLC.
- Translational pathway validation is ongoing.

REFERENCES

- Liu X, Bao X, Hu M, et al. Inhibition of PCSK9 potentiates immune checkpoint therapy for cancer. *Nature*. 2020;588(7839):693-698.