Establishment of patient-derived organoids from EBUS-TBNA cytological samples in lung cancer patients

P3.03.10

Melissa Acosta-Plasencia[1], Juan Pablo Orozco[2], Carmen Lucena[2], Yangyi He[1], Antonio Altuna-Coy[1], Risha Na[1], Tania Díaz[1], Yi Liu[1], Carles Agustí[2], Ramón M. Marrades[2,3], Alfons Navarro[1,4]

[1]Laboratory of Molecular Oncology and Embryology, Department of Surgery and Medical-Surgical Specialties, Unit of Human Anatomy and Embryology, Faculty of Medicine and Health Sciences, Casanova St. 143, 08036 Barcelona, Spain; [2]Department of Pulmonology, Clinical Respiratory Institute (ICR), Hospital Clínic de Barcelona, University of Barcelona, 08036 Barcelona, Spain; [3]Biomedical Research Networking Center in Respiratory Diseases (CIBERES), Instituto de Salud Carlos III, 28029 Madrid, Spain; [4]August Pi i Sunyer Biomedical Research Institute (IDIBAPS), Villarroel St. 170, 08036 Barcelona, Spain.

1. INTRODUCTION

Most lung cancer organoids (LCOs) are generated from surgical samples, limiting their use to resected patients (mostly early stage). This study explores the feasibility of establishing LCOs from lymph node metastases (LN-LCOs) obtained via EBUS-TBNA and assessed whether these organoids retain patient-specific histopathological features, potentially enabling their use as a complementary diagnostic tool.

2. METHODS

Cytological samples were obtained via EBUS-TBNA from 13 lung cancer patients (Table 1).

Table1. Patient's main clinic-pathological characteristics.

Characteristics	Value	N (%)
Sex	Male	7 (54%)
	Female	6 (46%)
Age (yrs)	Mean [range]	67 [52–84]
	≤65	7 (54%)
	>65	6 (46%)
Stage	I-II	1 (8%)
	III	6 (46%)
	IV	6 (46%)
Histology	Adenocarcinoma	7 (54%)
	Squamous cell carcinoma	2 (15%)
	Pleomorphic carcinoma	1 (8%)
	Small cell carcinoma	3 (23%)
Relapse	No	11 (85%)
	Yes	2 (15%)

The presence of tumor cells in all samples was confirmed by ROSE. Samples were enzymatically dissociated and cultured in a LCOs-enriched medium (A).

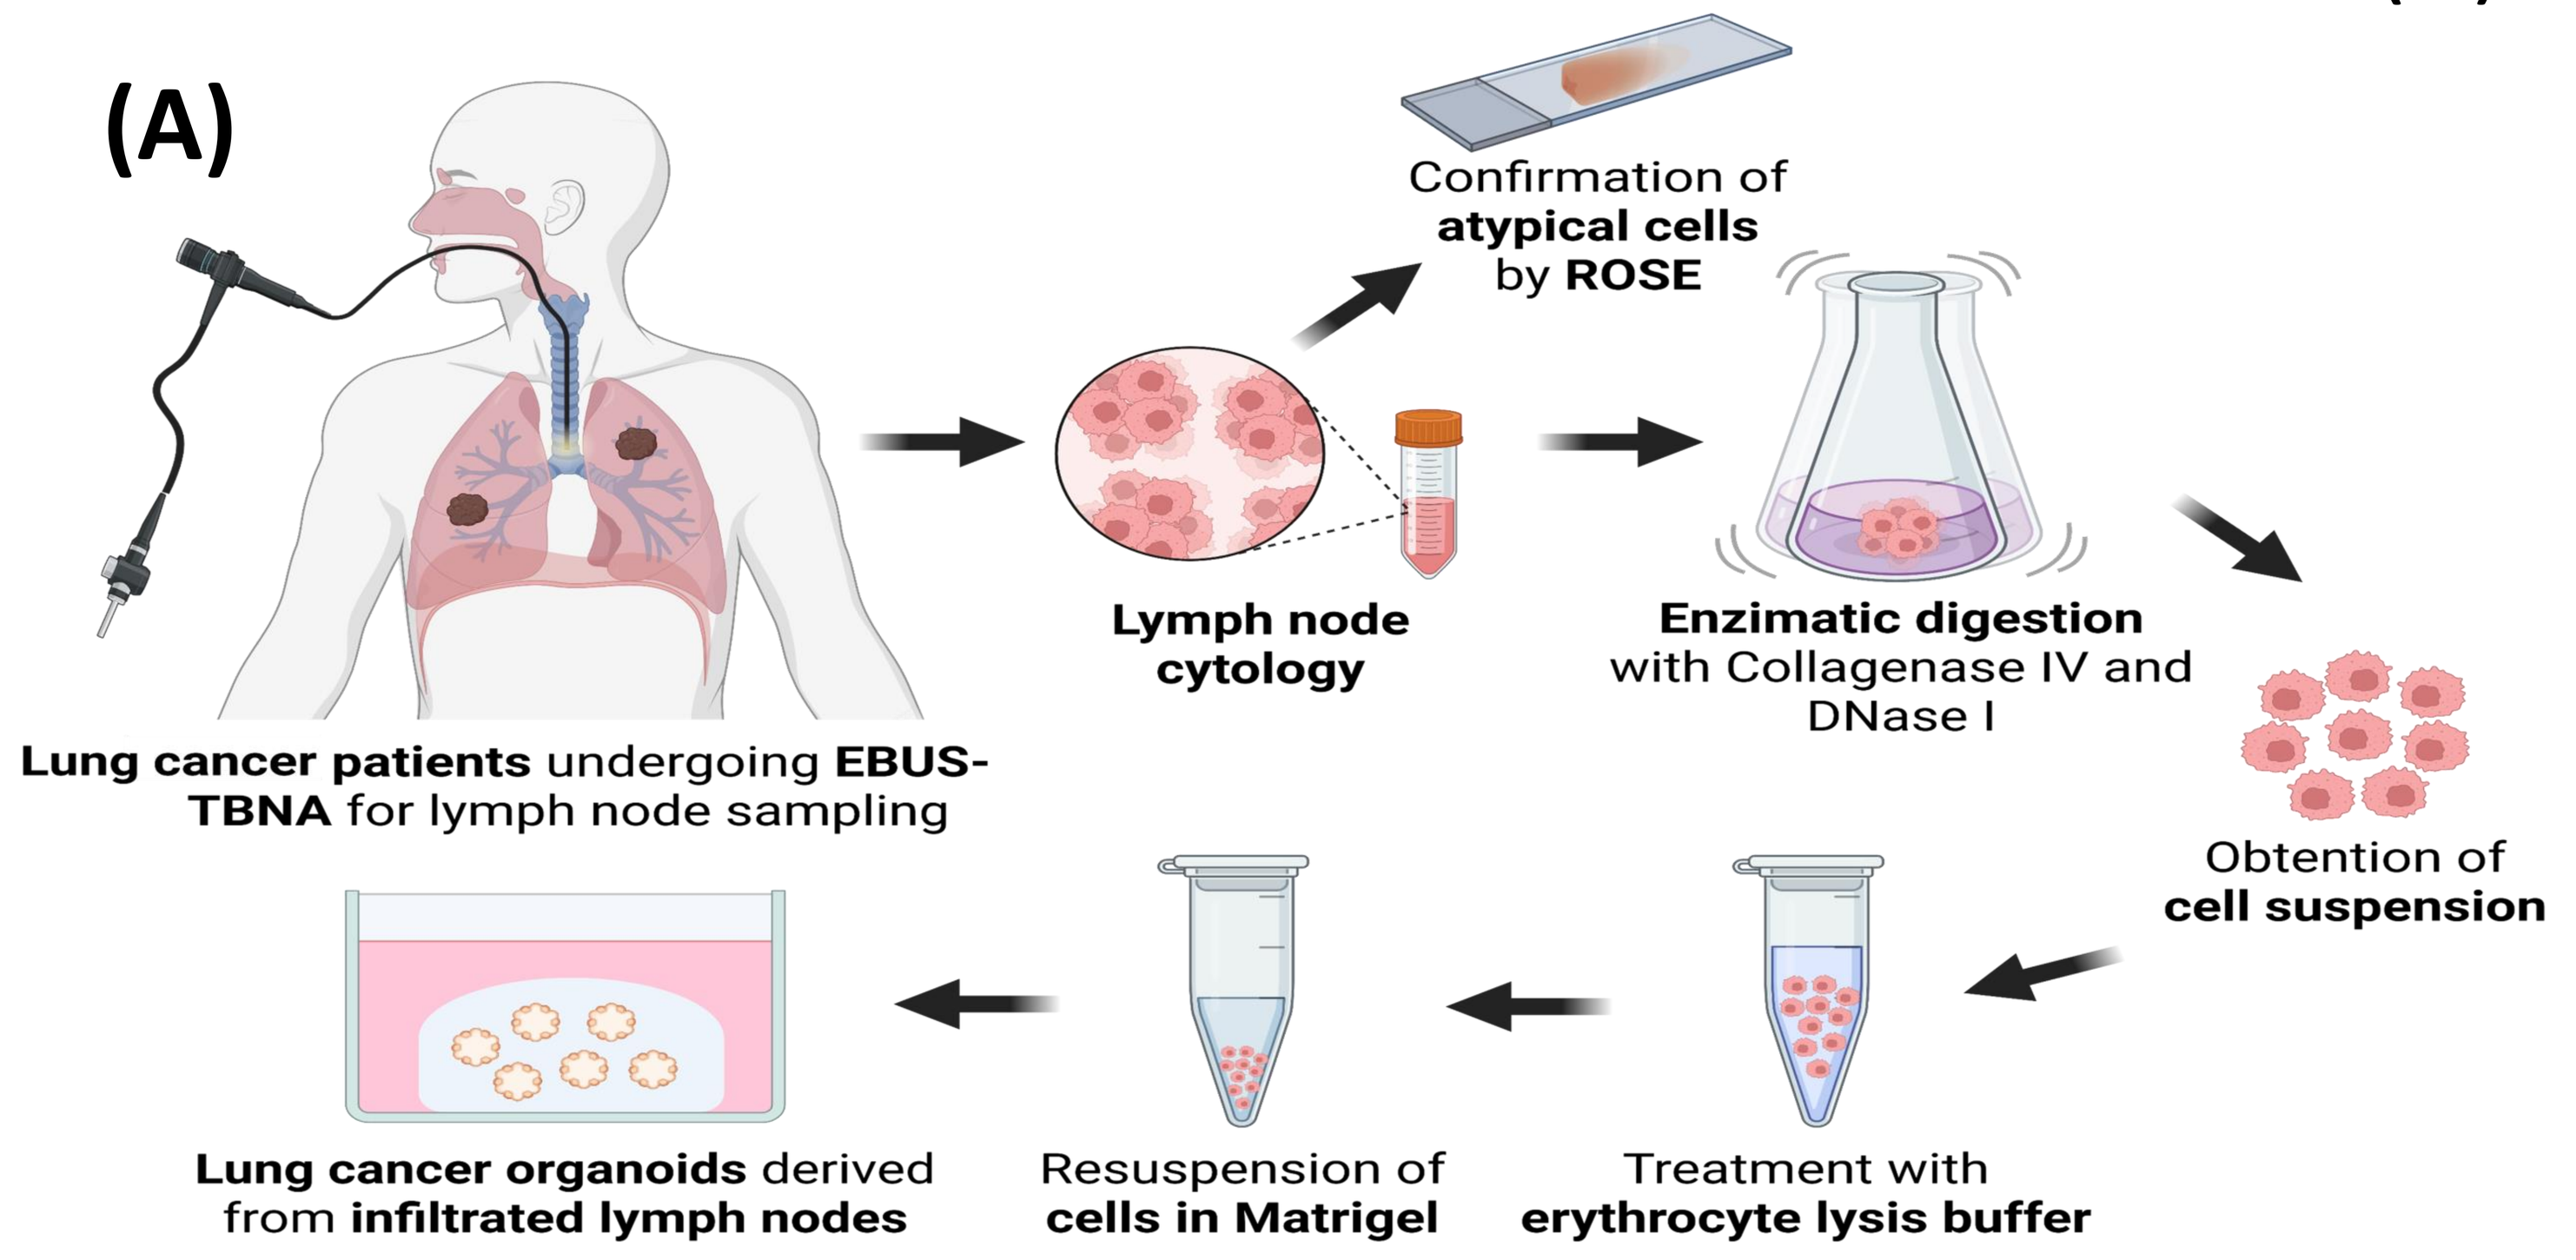
(A)
Confirmation of atypical cells by ROSE
Lung cancer patients undergoing EBUS-TBNA for lymph node sampling
Lymph node cytology
Enzimatic digestion with Collagenase IV and DNase I
Obtention of cell suspension
Lung cancer organoids derived from infiltrated lymph nodes
Resuspension of cells in Matrigel
Treatment with erythrocyte lysis buffer

3. RESULTS

1. LN-LCOs culture establishment. (B) Overall culture success rate. Representative image of a (C) failed and (D) successful culture. Success rate by (E) lung cancer type: non-small cell LC (NSCLC) vs. small cell LC (SCLC) (SCC); (F) histological subtype: adenocarcinoma (ADK) vs. squamous cell carcinoma; and (G) metastatic status. Successful Failed

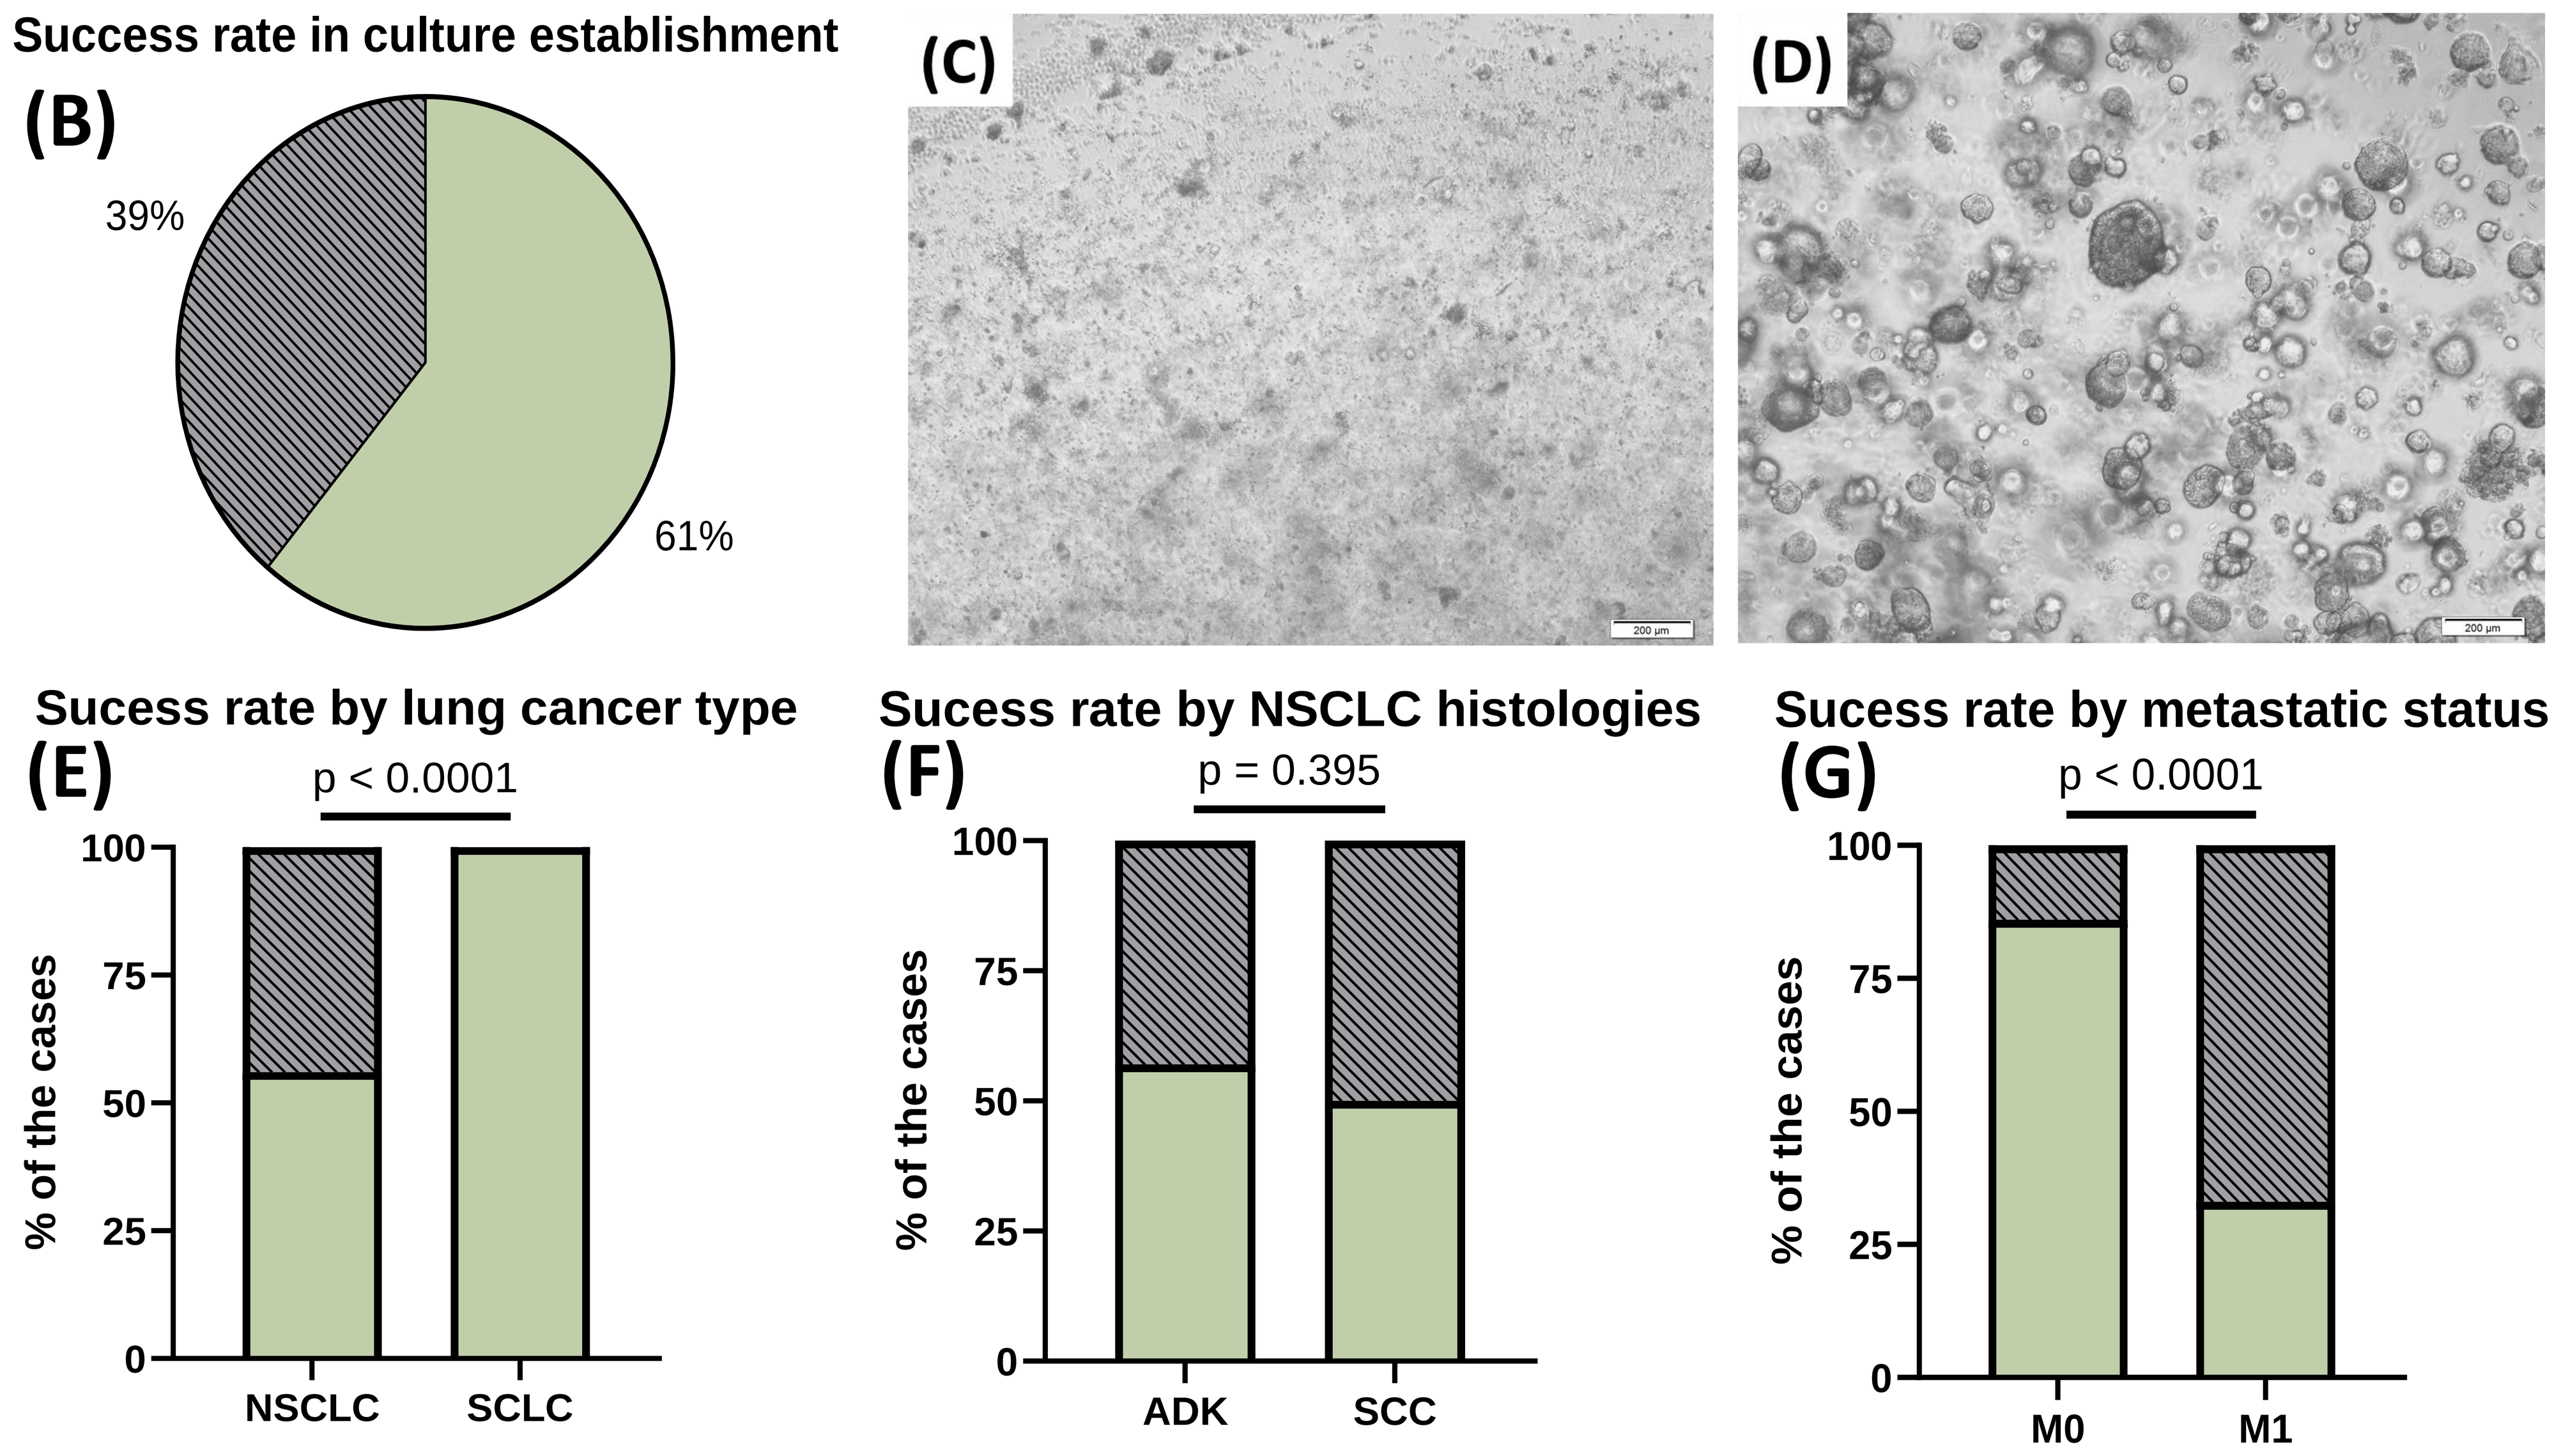
Success rate in culture establishment
(B)
39%
61%
(C)
(D)
Sucess rate by lung cancer type
(E)
p < 0.0001
% of the cases
100
75
50
25
0
NSCLC
SCLC
Sucess rate by NSCLC histologies
(F)
p = 0.395
% of the cases
ADK
SCC
Sucess rate by metastatic status
(G)
p < 0.0001
% of the cases
M0
M1

2. LN-LCOs recapitulate patient histopathological features. Representative images of hematoxylin and eosin staining of (H) an ADK-derived organoid and (I) a SCC-derived organoid; and positive immunohistochemical staining for (J) TTF-1 in an ADK-derived organoid and (K) P40 in a SCC-derived organoid.

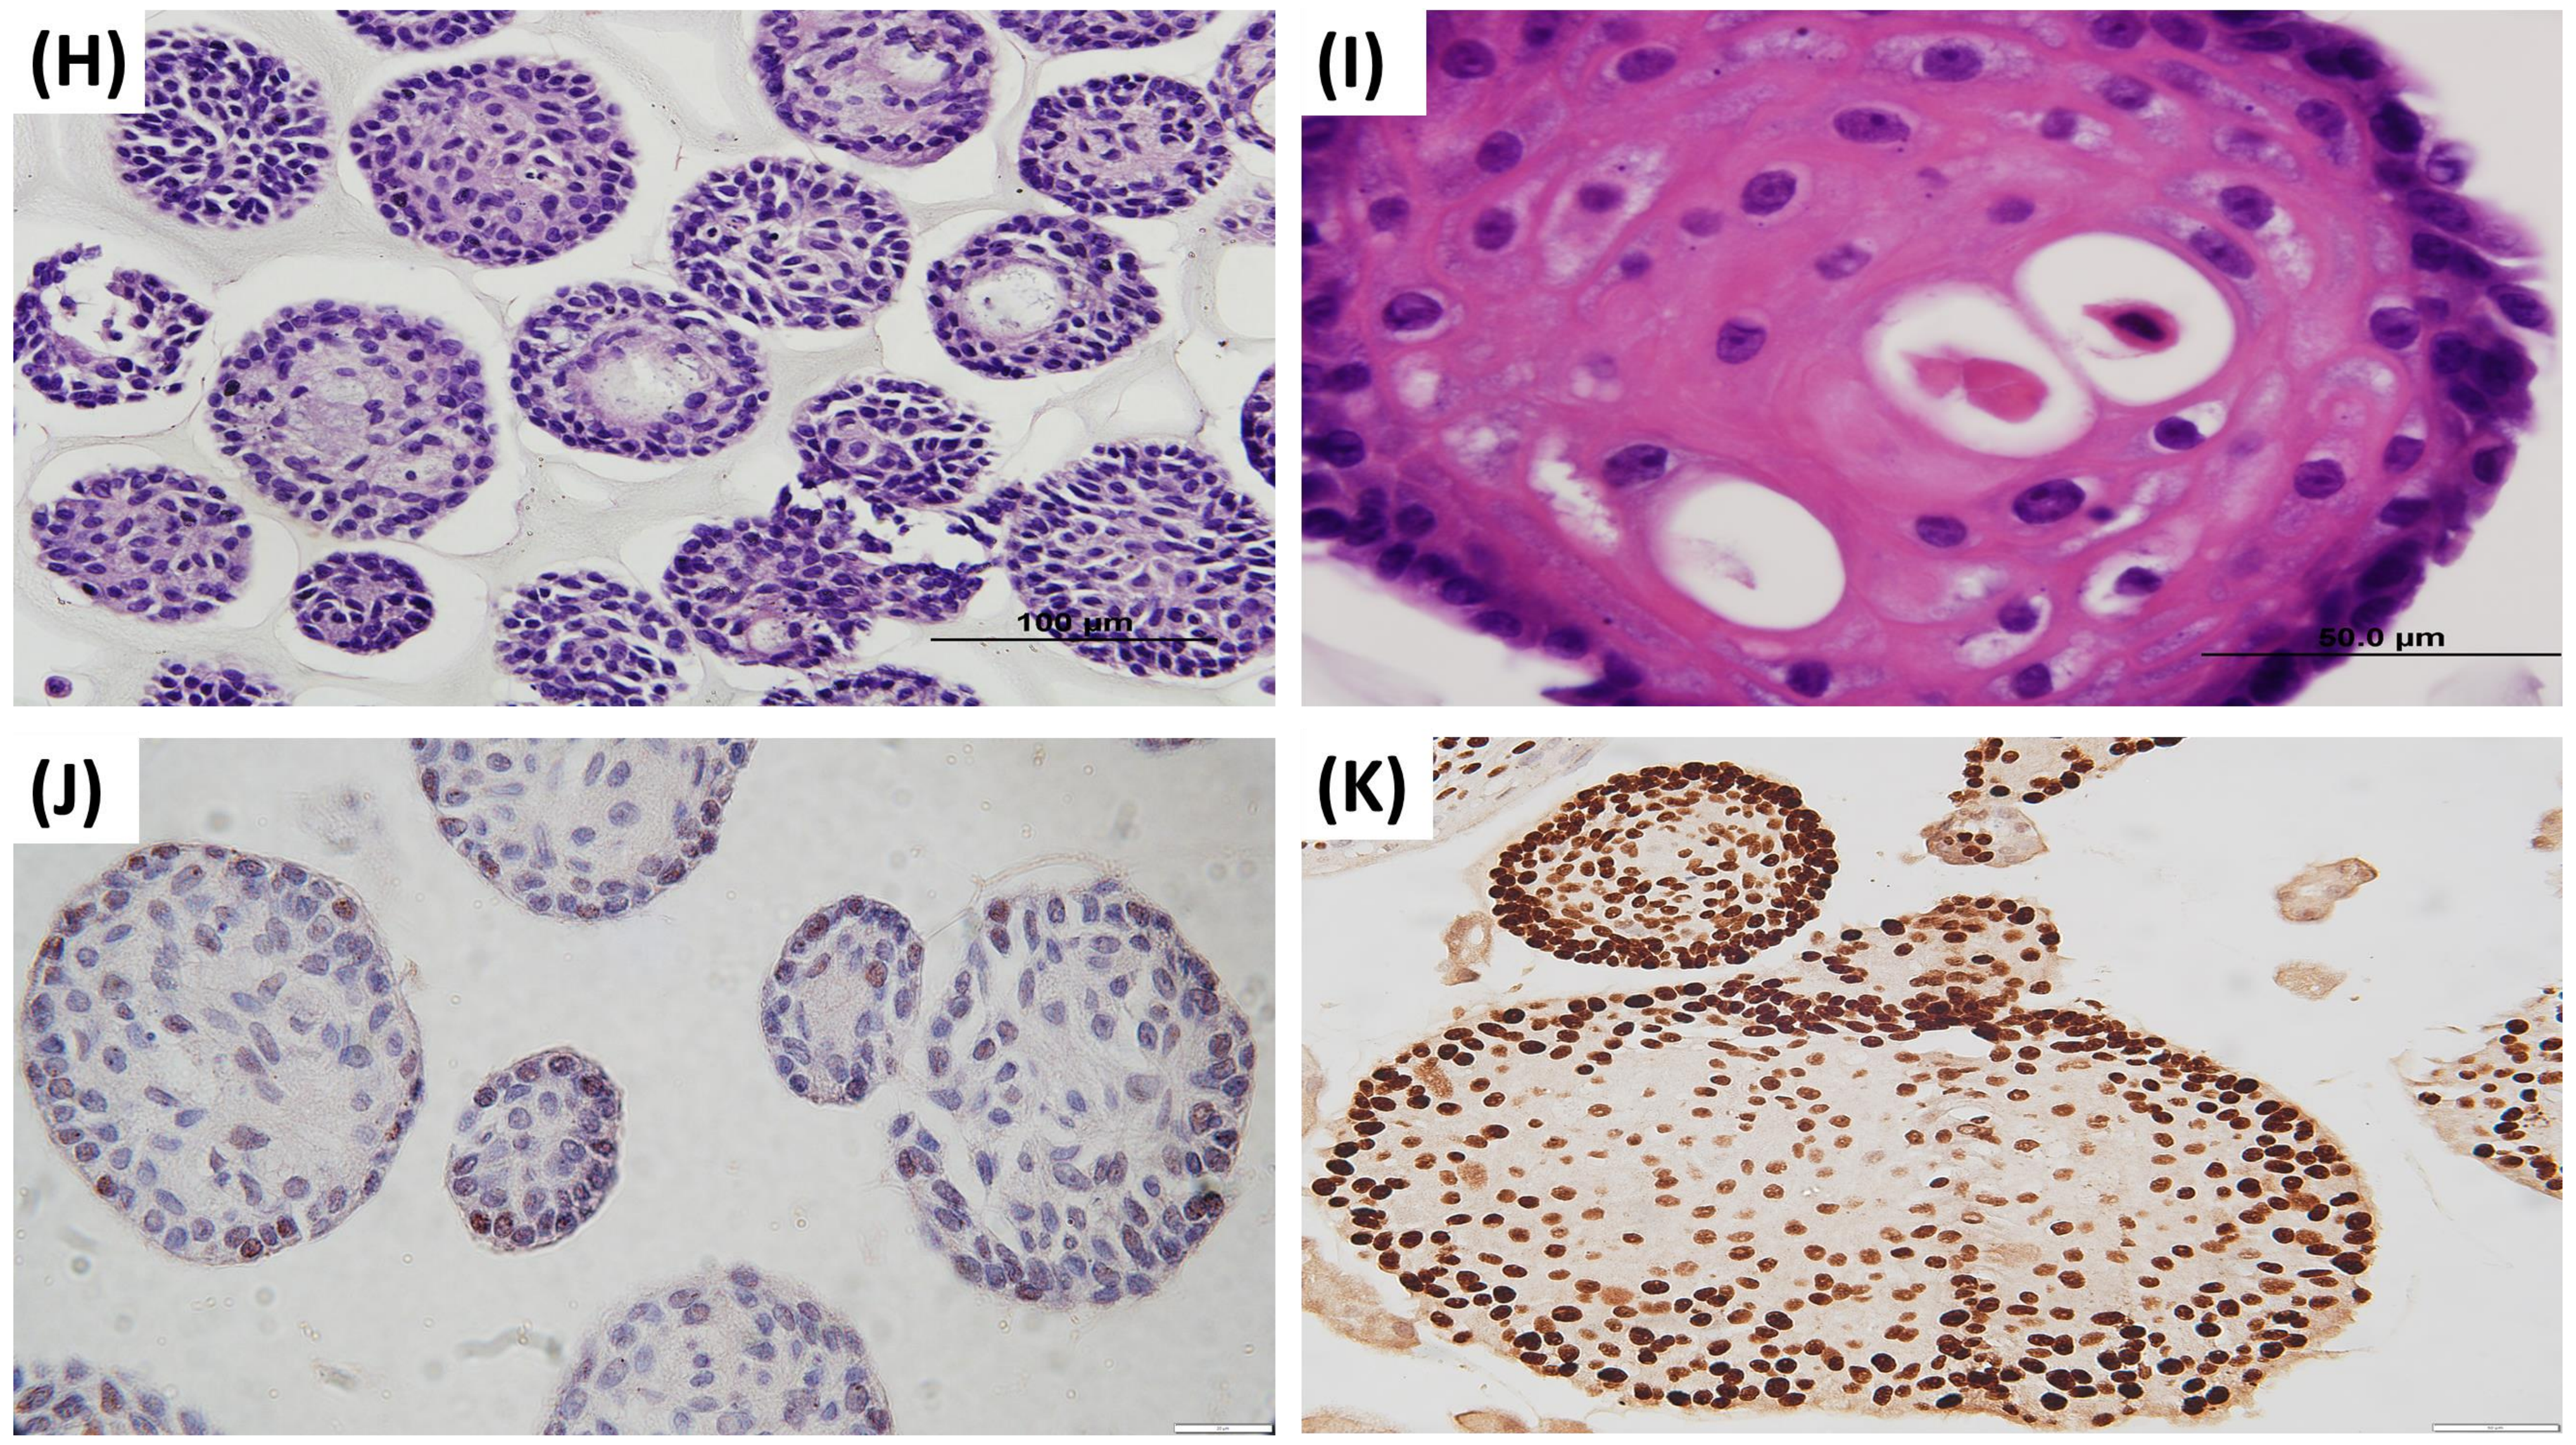
(H)
100 µm
(I)
50.0 µm
(J)
(K)

3. LN-LCOs show increased expression of mesenchymal markers. Comparison of gene expression between organoids derived from primary lung tumors (primary-LCOs) and LN-LCOs). CDH1 expression (L) showed no significant difference between groups, while SNAI1 (M) and ZEB1 (N), were significantly upregulated in LN-LCOs.

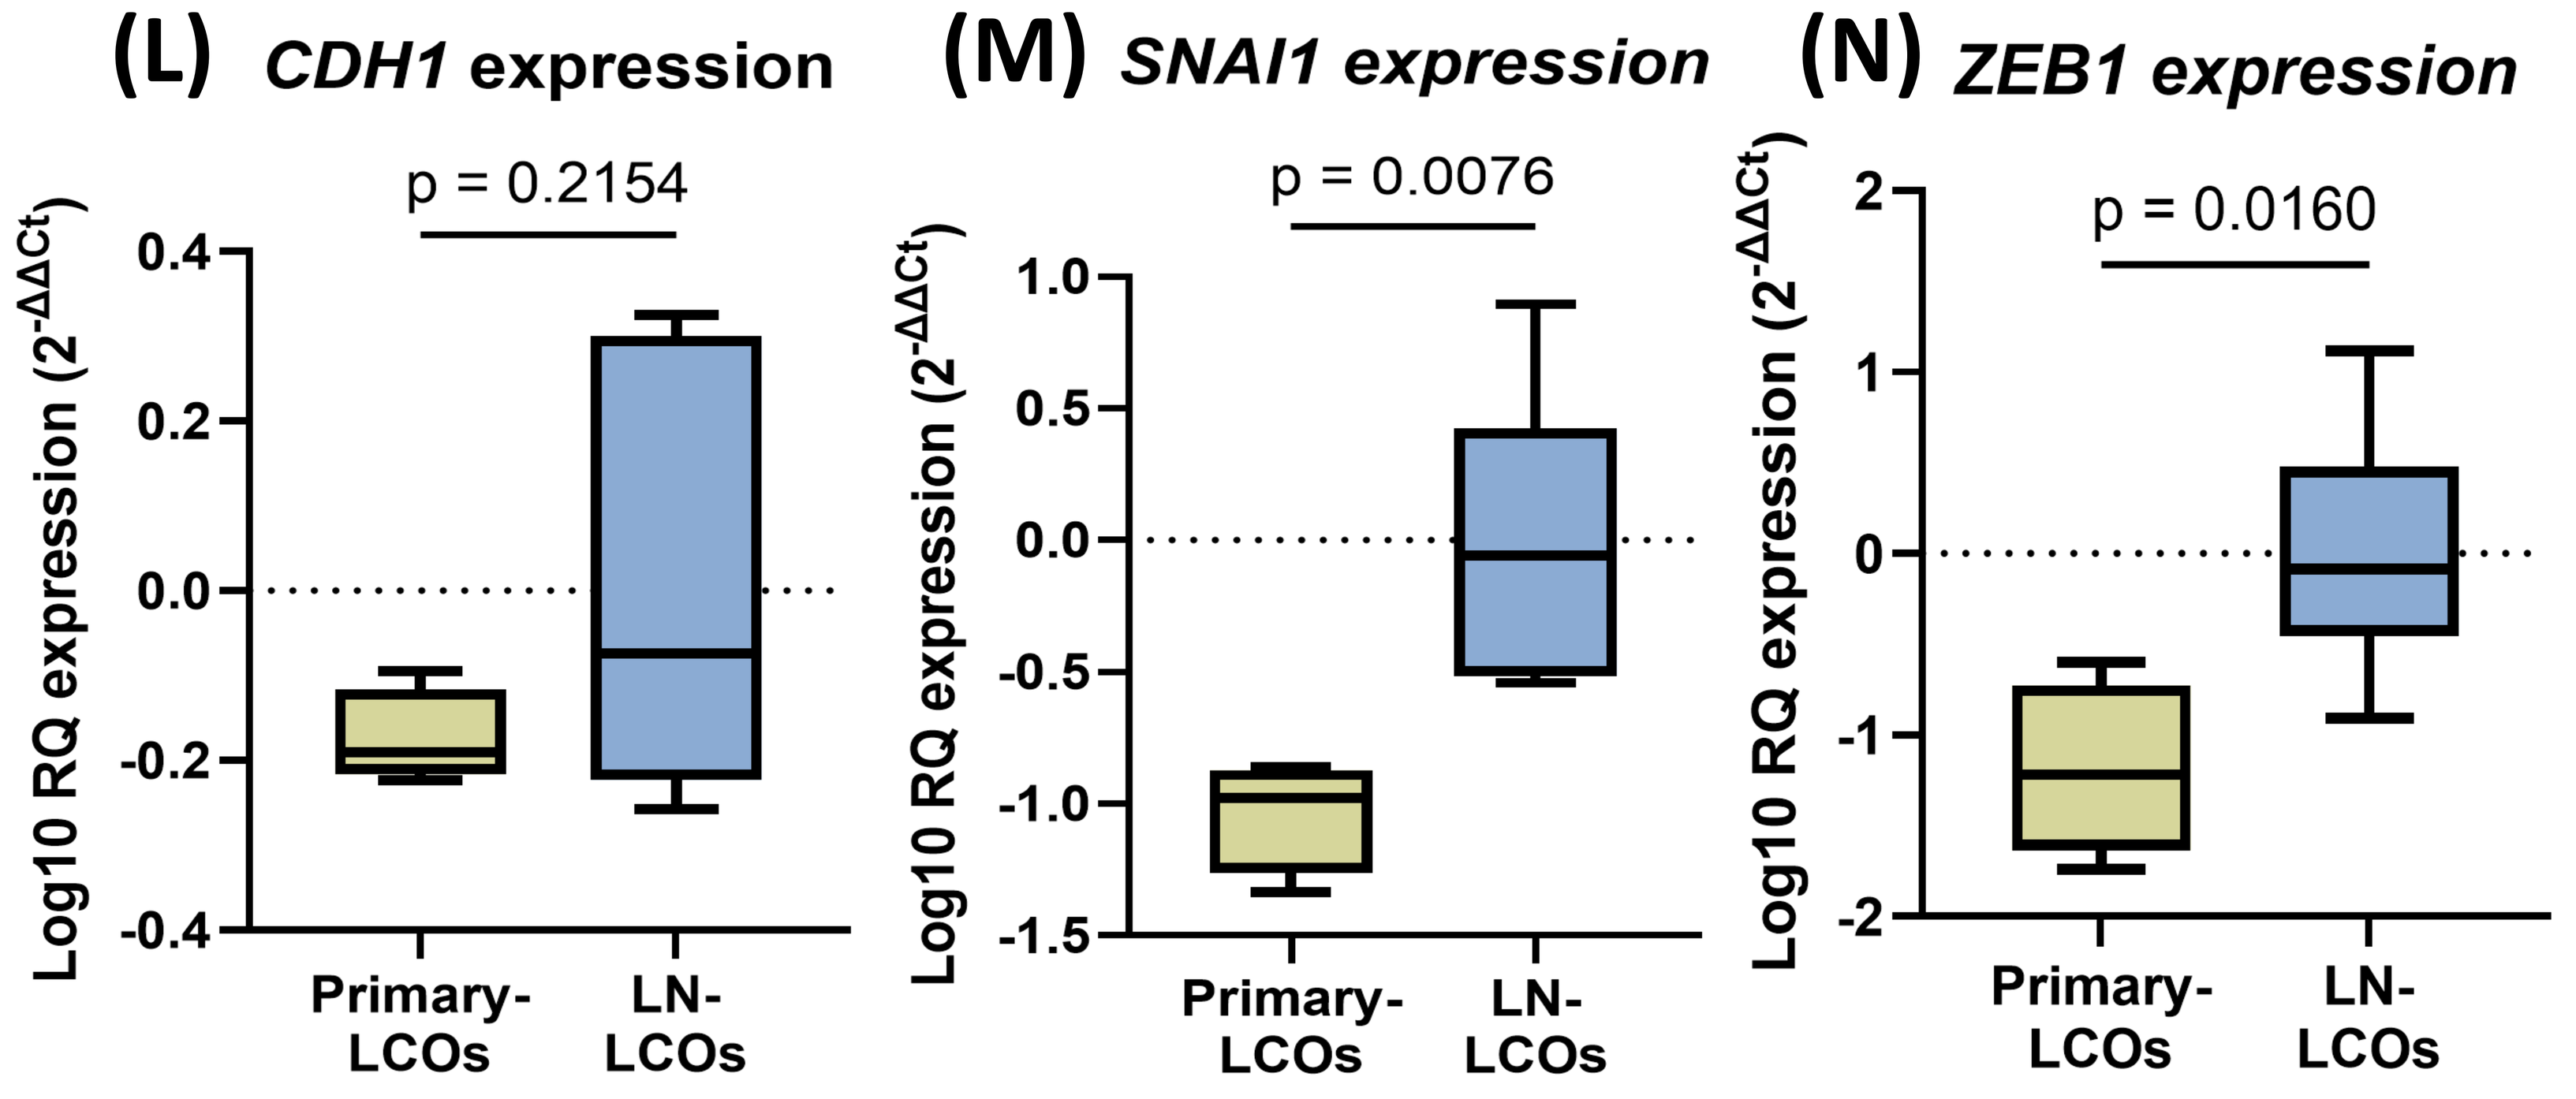
(L) CDH1 expression
p = 0.2154
(M) SNAI1 expression
p = 0.0076
(N) ZEB1 expression
p = 0.0160
Log10 RQ expression (2-ΔΔCt)
Primary-LCOs
LN-LCOs

4. CONCLUSIONS

LN-LCOs derived from EBUS-TBNA samples replicate the patient's tumor histopathology, offering a novel strategy to optimize sample use and enable studies in advanced-stage lung cancer patients.

5. REFERENCES AND FUNDINGS

Acosta-Plasencia M., et al (2022). Patient-derived organoids as a stem cell model to study lung cancer. *Interdisciplinary Cancer Research: Springer*. p. 319-346.

Acosta-Plasencia M., et al (2024). Potential of Patient-Derived Organoids and Other 3D Culture Models to Assess Immunotherapy Response in Cancer. *Handbook of Cancer and Immunology: Springer*. p. 1-29.

Acosta-Plasencia, M., et al (2025). Selection of the most suitable culture medium for patient-derived lung cancer organoids. *Cells Tissues Organs*, 214(2), 114–127.

This work was supported by the 2021 Sociedad Española de Neumología y Cirugía Torácica (SEPAR)–AstraZeneca PII Oncology Research Grant (no. 003); the 2022 SEPAR Research Grant (no. 1326); the 2023 SEPAR Research Grant (no. 1482); and the 2025 Fellowship Grant from the SEPAR.